Poster ID:
Wed-PT-38

Reduced Pain and Regional Brain Activation following Tetrodotoxin (TTX) Treatment in a Cynomolgus Model of Post-operative Pain

D. Wong[1], W. Korz[1], Y. Awaga[2], M. Yano[2], R. Fujii[2], T. Natsume[2], A. Hama[2], H. Takamatsu[2]

[1] WEX Pharmaceuticals Inc., Vancouver, Canada, [2] Hamamatsu Pharma Research Inc, Hamamatsu, Japan

Tetrodotoxin

Tetrodotoxin (TTX), trademark Halneuron®, is a small molecule that blocks voltage-gated sodium channels on peripheral neurons.

It exerts its analgesic effect by inhibiting the initiation and conduction of impulses in the nervous system.

TTX does not cross the blood-brain barrier. Mechanisms of action are exclusively peripheral.

Clinical trials have been ongoing to evaluate the analgesic effect of TTX in chemotherapy induced neuropathic pain, cancer pain, as well as other neuropathic and nociceptive pain conditions.

Specific Aim

To examine by fMRI how the peripheral actions of TTX is reflected in the CNS established pain regions in a macaque post-operative pain model.

Methods & Materials

Cynomolgus macaque model of postoperative pain

Following baseline assessment, under isoflurane anesthesia, a midline 3 cm incision was made on the epigastric region, above the umbilicus. Using aseptic technique, an incision was made through the skin and muscle. The wound was closed in layers and macaques were observed for recovery.

For acute post-op pain, one dose of buprenorphine (0.03 mg/kg, i.m.) was administered, within 10 min., after the surgery (Day 0). Enrofloxacin (5 mg/kg, i.m.) was administered once daily for three days.

Treatment with TTX or vehicle

Treatment	Dose	Schedule	Route	n
Vehicle (saline)	0.1mL/kg	Day 1 pm, Day 2 am & pm, Day 3 am	SC	6
TTX	2 µg/kg	Day 1 pm, Day 2 am & pm, Day 3 am	SC	6

Algometer-pressure test was performed in the morning of Day 1 before TTX to determine baseline, then in the afternoon of Day 1, morning and afternoon of Day 2, and morning of Day 3 about 1hour after TTX or vehicle administration.

fMRI was performed in the morning of Day1 after the pressure test but before randomization, as well as in the afternoon of Day 3 after the pressure test.

Algometer-Pressure Test

Response to pressure applied to the abdomen proximal (adjacent to the incision) and distal (about 10 cm) to the surgical incision site was measured with a modified pressure algometer. Average of three measurements was used.

fMRI

Propofol sedated animals were fixed on a MRI bed and brain activation was assessed using a 3.0T MRI system (Signa HDxt 3.0T MRI system (GE Healthcare, Milwaukee, WI, USA)).

During fMRI scan, animals underwent a block design stimulation protocol: 10 sets of abdominal pressure stimulations. One stimulation set consisted of 30 sec. of an "OFF" stimulus, a 100 g weight (empty 1.5 L water bottle) applied by hand to rest perpendicularly on the skin, followed by 30 sec. of an "ON" stimulus, a 1.5 kg weight (1.5 L water bottle filled with water) applied by hand to rest perpendicularly on the skin.

Results

Functional Magnetic Resonance Imaging (fMRI)

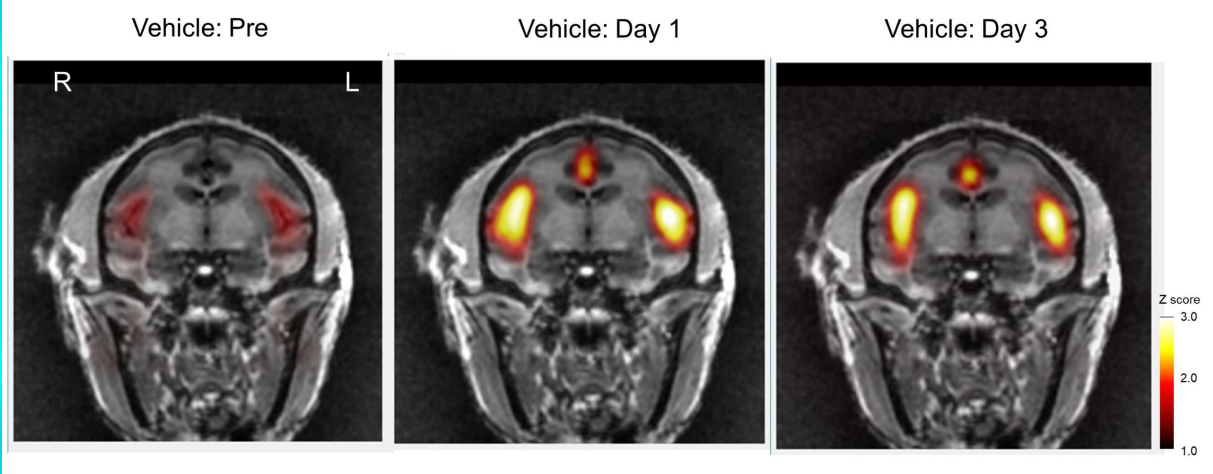
Vehicle: Pre
Vehicle: Day 1
Vehicle: Day 3
R
L
Z score
3.0
2.0
1.0

Figure 1. Vehicle treatment. Prior to incisional surgery, no significant activation was observed with non-noxious stimulation of the abdominal area. 1 day after incisional surgery, but before saline treatment, pressure evoked activation of the insular/secondary somatosensory cortex (Ins/SII) and cingulate cortex (CC). Robust pressure activation was also observed 3 days after surgery.

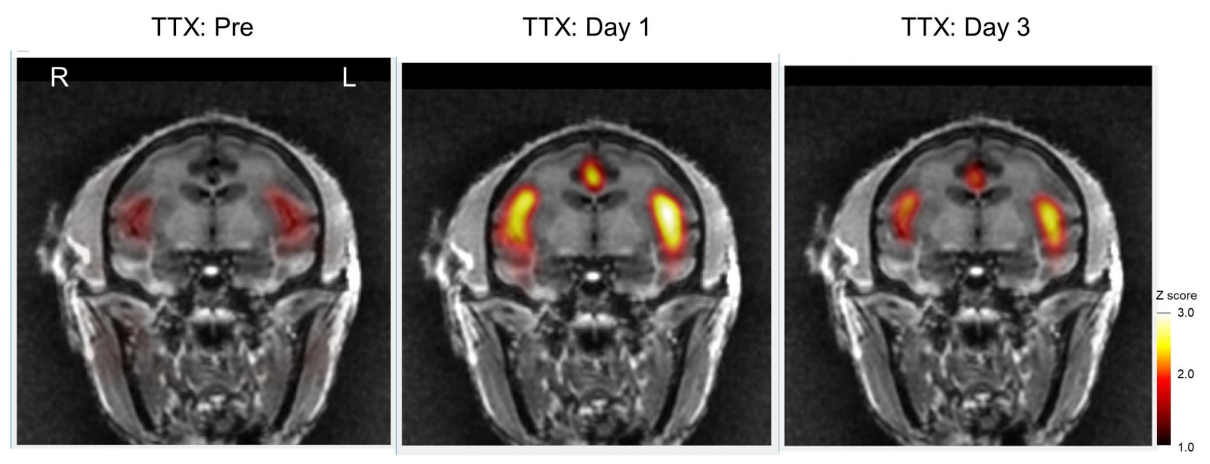
TTX: Pre
TTX: Day 1
TTX: Day 3
R
L
Z score
3.0
2.0
1.0

Figure 2. TTX treatment. Prior to incisional surgery, no significant activation was observed with non-noxious stimulation. One day after surgery (Day 1), but before TTX, pressure evoked activation of the Ins/SII and CC. Three days after surgery (Day 3) and TTX, reduced pressure activation.

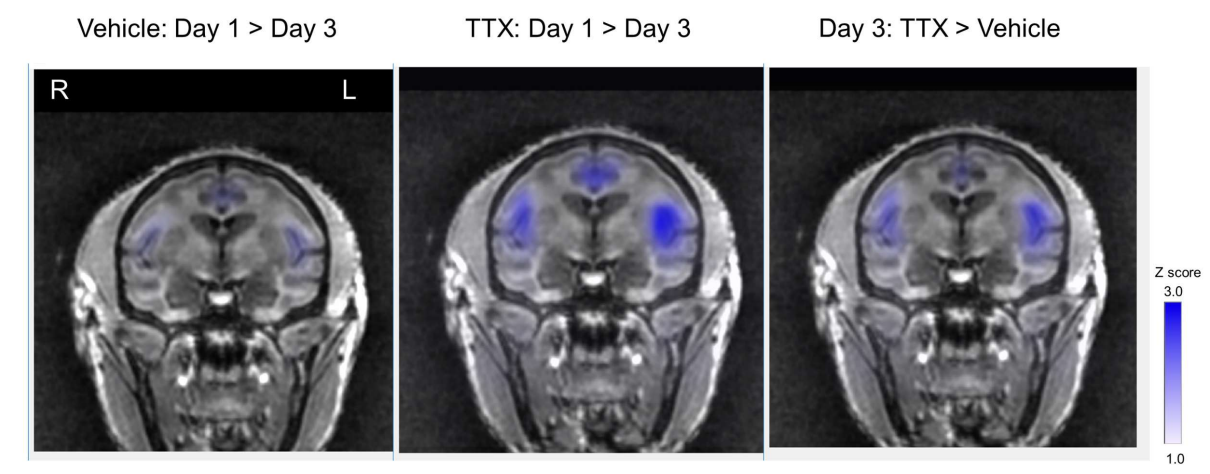
Vehicle: Day 1 > Day 3
TTX: Day 1 > Day 3
Day 3: TTX > Vehicle
R
L
Z score
3.0
1.0

Figure 3. Contrast maps: comparisons within treatments between Day 1 and Day 3 and comparison between treatments at Day 3. Following saline treatments, no changes in non-noxious pressure activation of the Ins/SII and CC were observed (Vehicle: Day 1 > Day 3). Following TTX treatments, reduced activation, or "deactivation", of the right Ins/SII was observed (TTX: Day 1 > Day 3). Comparing TTX and vehicle-treated macaques on Day 3, "deactivation" of the right Ins/SII was observed (Day 3: TTX > Vehicle).

Vehicle: Day 1 > Day 3

Area	Hemisphere	z-score	Coordinates (mm) x	y	z
Ins/SII	Left	0.41	-20	18	2
	Right	0.34	20	16	2
CC		0.28	-2	24	2

TTX: Day 1 > Day 3

Area	Hemisphere	z-score	Coordinates (mm) x	y	z
Ins/SII	Left	1.42	-20	16	2
	Right	2.81 **	18	16	2
CC		1.51	-2	22	2

Day 3: TTX > Vehicle

Area	Hemisphere	z-score	Coordinates (mm) x	y	z
Ins/SII	Left	1.13	-20	16	2
	Right	2.34 **	22	16	2
CC		0.33	-2	22	2

Table 1. Contrast maps: comparisons of peak voxel z-scores between Day 1 and Day 3 and between treatments on Day 3. No significant differences in peak voxels were observed between Day 1 and Day 3 in vehicle-treated macaques with a surgical incision. No significant differences in peak voxels of the left Ins/SII and CC were observed between Day 1 and Day 3 in TTX-treated macaques with a surgical incision. However, "deactivation" was observed in the right Ins/SII (z-score greater than 2.3, P < 0.01). Furthermore, comparing TTX-treated and vehicle-treated macaques on Day 3, "deactivation" of the right Ins/SII was observed. Peak activation voxels z-scores are shown. Z-scores greater than 2.3, ** P < 0.01. Values are averages from scans obtained from 6 macaques per treatment group.

Algometer-Pressure Test

*P < 0.05 TTX vs vehicle on Day 2, AM.

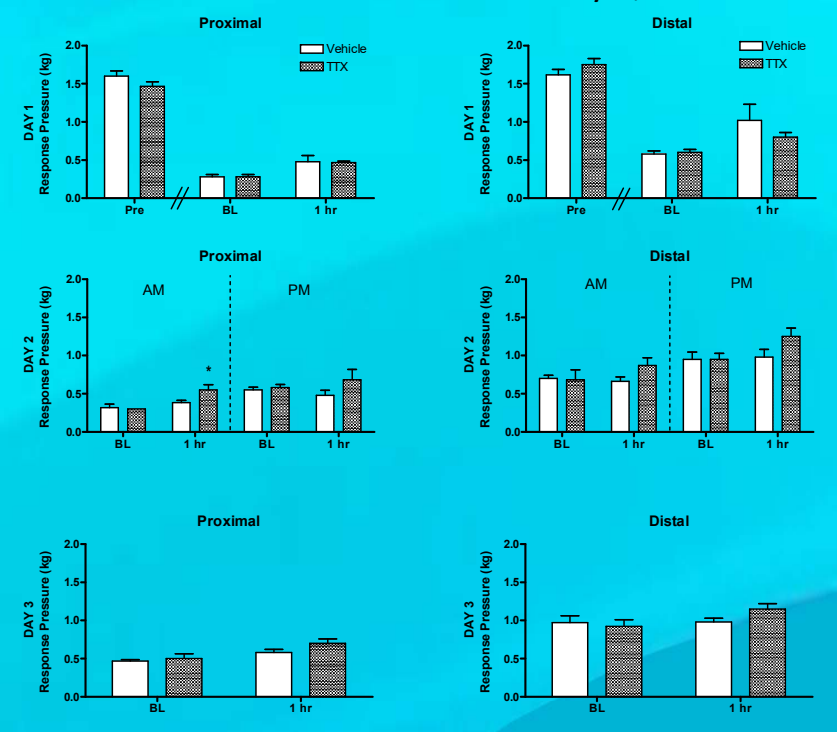
Proximal
Distal
Vehicle
TTX
DAY 1
DAY 2
DAY 3
Response Pressure (kg)
2.0
1.5
1.0
0.5
0.0
Pre
BL
1 hr
AM
PM
*

Figure 4. Response pressures (kg) were measured prior to surgery (Pre). Twenty-four hours following surgery (Day 1), response pressures were measured (BL), proximally and distally to the incision, prior to s.c. treatment of either vehicle or TTX. Response pressures were measured one hour after treatment. On Day 2, macaques received a morning and afternoon dose of either vehicle or TTX. On Day 3, macaques received one morning dose. Data are expressed as mean ± SEM. N = 6 per group. *P < 0.05

Summary and Conclusion

- It is well documented and we had previously shown that TTX acts peripherally and does not cross the BBB.
- Analgesia from the action of TTX on the PNS may be reflected in the CNS as "deactivation" of the right Ins/SII.

Relevance

- The literature shows that of all the areas involved in pain, only the Ins and SII generate pain when stimulated.
- Ins lesions lead to nociceptive deficits.
- Activation of Ins/SII and CC have been observed in patients with neuropathic pain.
- The Ins is extensively connected and has been suggested to modulate multiple components of pain, including motivational / affective, sensorimotor integration, and descending pain-inhibition.
- Nociceptive processing occurs within SII and may involve recognition, learning and memory of pain.
- Persistent changes in the Ins have been suggested to underlie chronic pain.
- Evoked brain activation could be utilized as a quantifiable pain biomarker of treatment efficacy.

The efficacy of Halneuron® (TTX) for chemotherapy-induced neuropathic pain is being further investigated in human phase II & III clinical trials.

Disclosures

This study was funded by WEX Pharmaceuticals Inc. DW and WK are employees of WEX. YA, MY, RF, TN, AH and HT are employees of Hamamatsu Pharma Research who performed the preclinical study on contract. All procedures in this study were approved by the Animal Care and Use Committee at Hamamatsu Pharma Research, Inc. and were conducted in accordance with the Guide for the Care and Use of Laboratory Animals (National Academy of Science, 2011). The facility where the study was conducted is accredited by the Association for the Assessment and Accreditation of Laboratory Animal Care International.