Tetrodotoxin Attenuates Thermal Hyperalgesia in a Rat Full Thickness Thermal Injury Model

Margaux M. Salas PhD[1], Donald Wong PhD[2], Walter Korz[2], John L. Clifford PhD[1]
United States Army Institute of Surgical Research, JBSA Fort Sam Houston, TX 78234-7677

Abstract

Burn injuries have been identified as the primary cause of injury in 5% U.S. military personnel evacuated from Operations Iraqi Freedom and Enduring Freedom. Severe burn-associated pain is typically treated with opioids such as fentanyl (94.1% of patients in ISR Burn unit), morphine (90.1%), and methadone (87.1%). Side effects of opioids include the development of hyperalgesia, addiction, inflammation, respiratory depression, nausea and sedation. These effects have led us to search for novel analgesics for the treatment of burn-associated pain in wounded warriors. Tetrodotoxin (TTX) is a selective voltage-gated sodium channel blocker currently in clinical trials as an analgesic for cancer-related pain and has been shown in mice to inhibit the development of neuropathic pain. TTX was originally identified as a neurotoxin in marine animals but has been shown to be safe in humans with chemotherapy-induced neuropathic pain at low doses. The antinociceptive effects of TTX are thought to be due to inhibition of Na+ ion influx required for initiation and conduction of nociceptive impulses. One TTX sensitive sodium channel, $Na_v1.7$, has been shown to be essential in lowering the heat pain threshold after burn injuries. To date, the analgesic effect of TTX has not been tested in burn-associated pain. Male Sprague-Dawley rats were subjected to a full thickness thermal injury on the right hind paw. TTX (8ug/kg) was administered daily, subcutaneously beginning at 3 days post thermal injury and continued through 7 days post thermal injury. Thermal hyperalgesia and mechanical allodynia were assessed 60 and 120 minutes post injection. TTX significantly reduced thermal hyperalgesia at all-time points tested. These results suggest that TTX may be an effective, rapidly acting analgesic for battlefield burn injuries.

TTX Binding sites on Na^+ channels

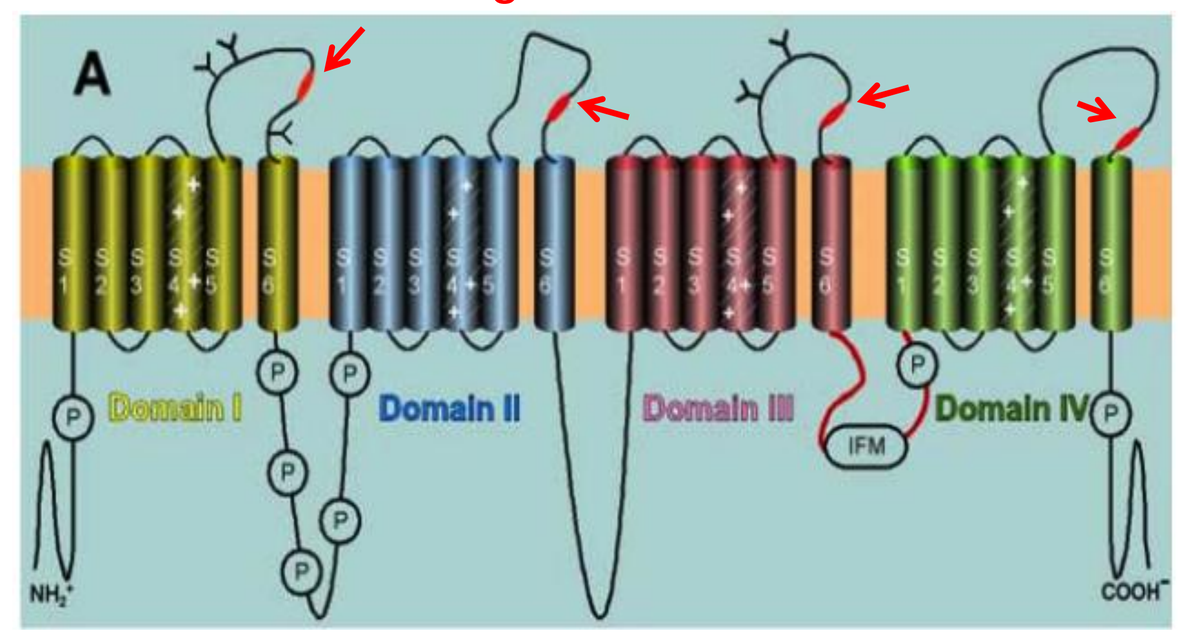
A
Domain I
Domain II
Domain III
Domain IV
IFM

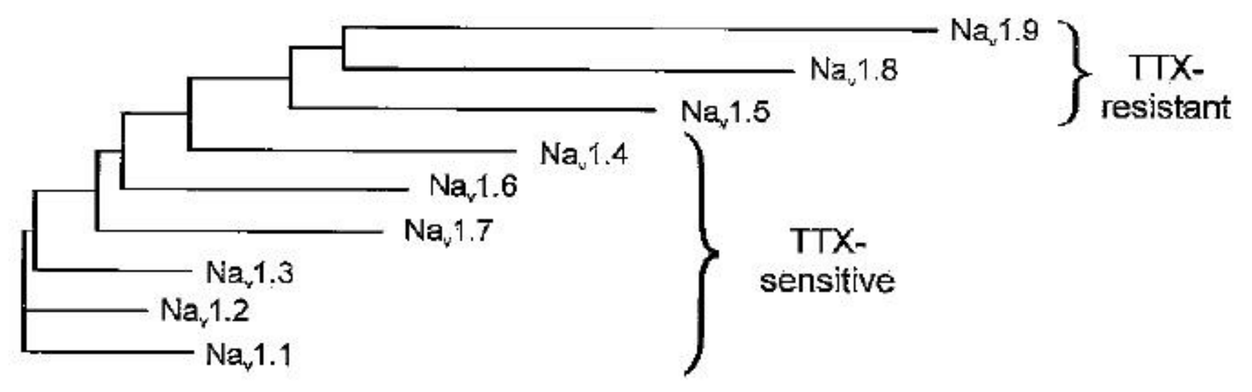
Na$_v$1.9
Na$_v$1.8
Na$_v$1.5
TTX-resistant
Na$_v$1.4
Na$_v$1.6
Na$_v$1.7
Na$_v$1.3
Na$_v$1.2
Na$_v$1.1
TTX-sensitive

Denac, H., Mevissen, M., & Scholtysik, G. (2000). Structure, function and pharmacology of voltage-gated sodium channels. Naunyn-Schmiedeberg's Archives of Pharmacology, 362(6), 453–479.

Beaulieu, P., David, L., Porreca, F., & Dickenson, A. H. (Eds.). (2010). *Pharmacology of Pain*. IASP Press, International Association for the Study of Pain

Methods

Full Thickness Thermal Injury

Male Sprague-Dawley rats, under general inhalation anesthesia, had a temperature-controlled, 100°C slanted soldering tip placed on the right hindpaw for 30 seconds to induce thermal injury.

Pain Behavior Testing

A paw thermal stimulator was used to assess paw withdrawal latencies (PWL) to a noxious thermal stimulus as a measure of *thermal hyperalgesia* (heightened sensitivity to a normally painful stimulus).

An electric anethesiometer was used to assess sensitivity to a mechanical stimulus as a measure of mechanical *allodynia* (sensitivity to a normally non-painful stimulus)

Rats were randomized to treatments and behavioral testing was conducted by an observer blinded to drug delivered. Rats received either morphine (5 mg/kg; S.C.; n = 9), TTX (8 µg/kg; S.C.; n = 9) or saline (S.C.; n = 9)

Rodent Full Thickness Thermal Injury Model

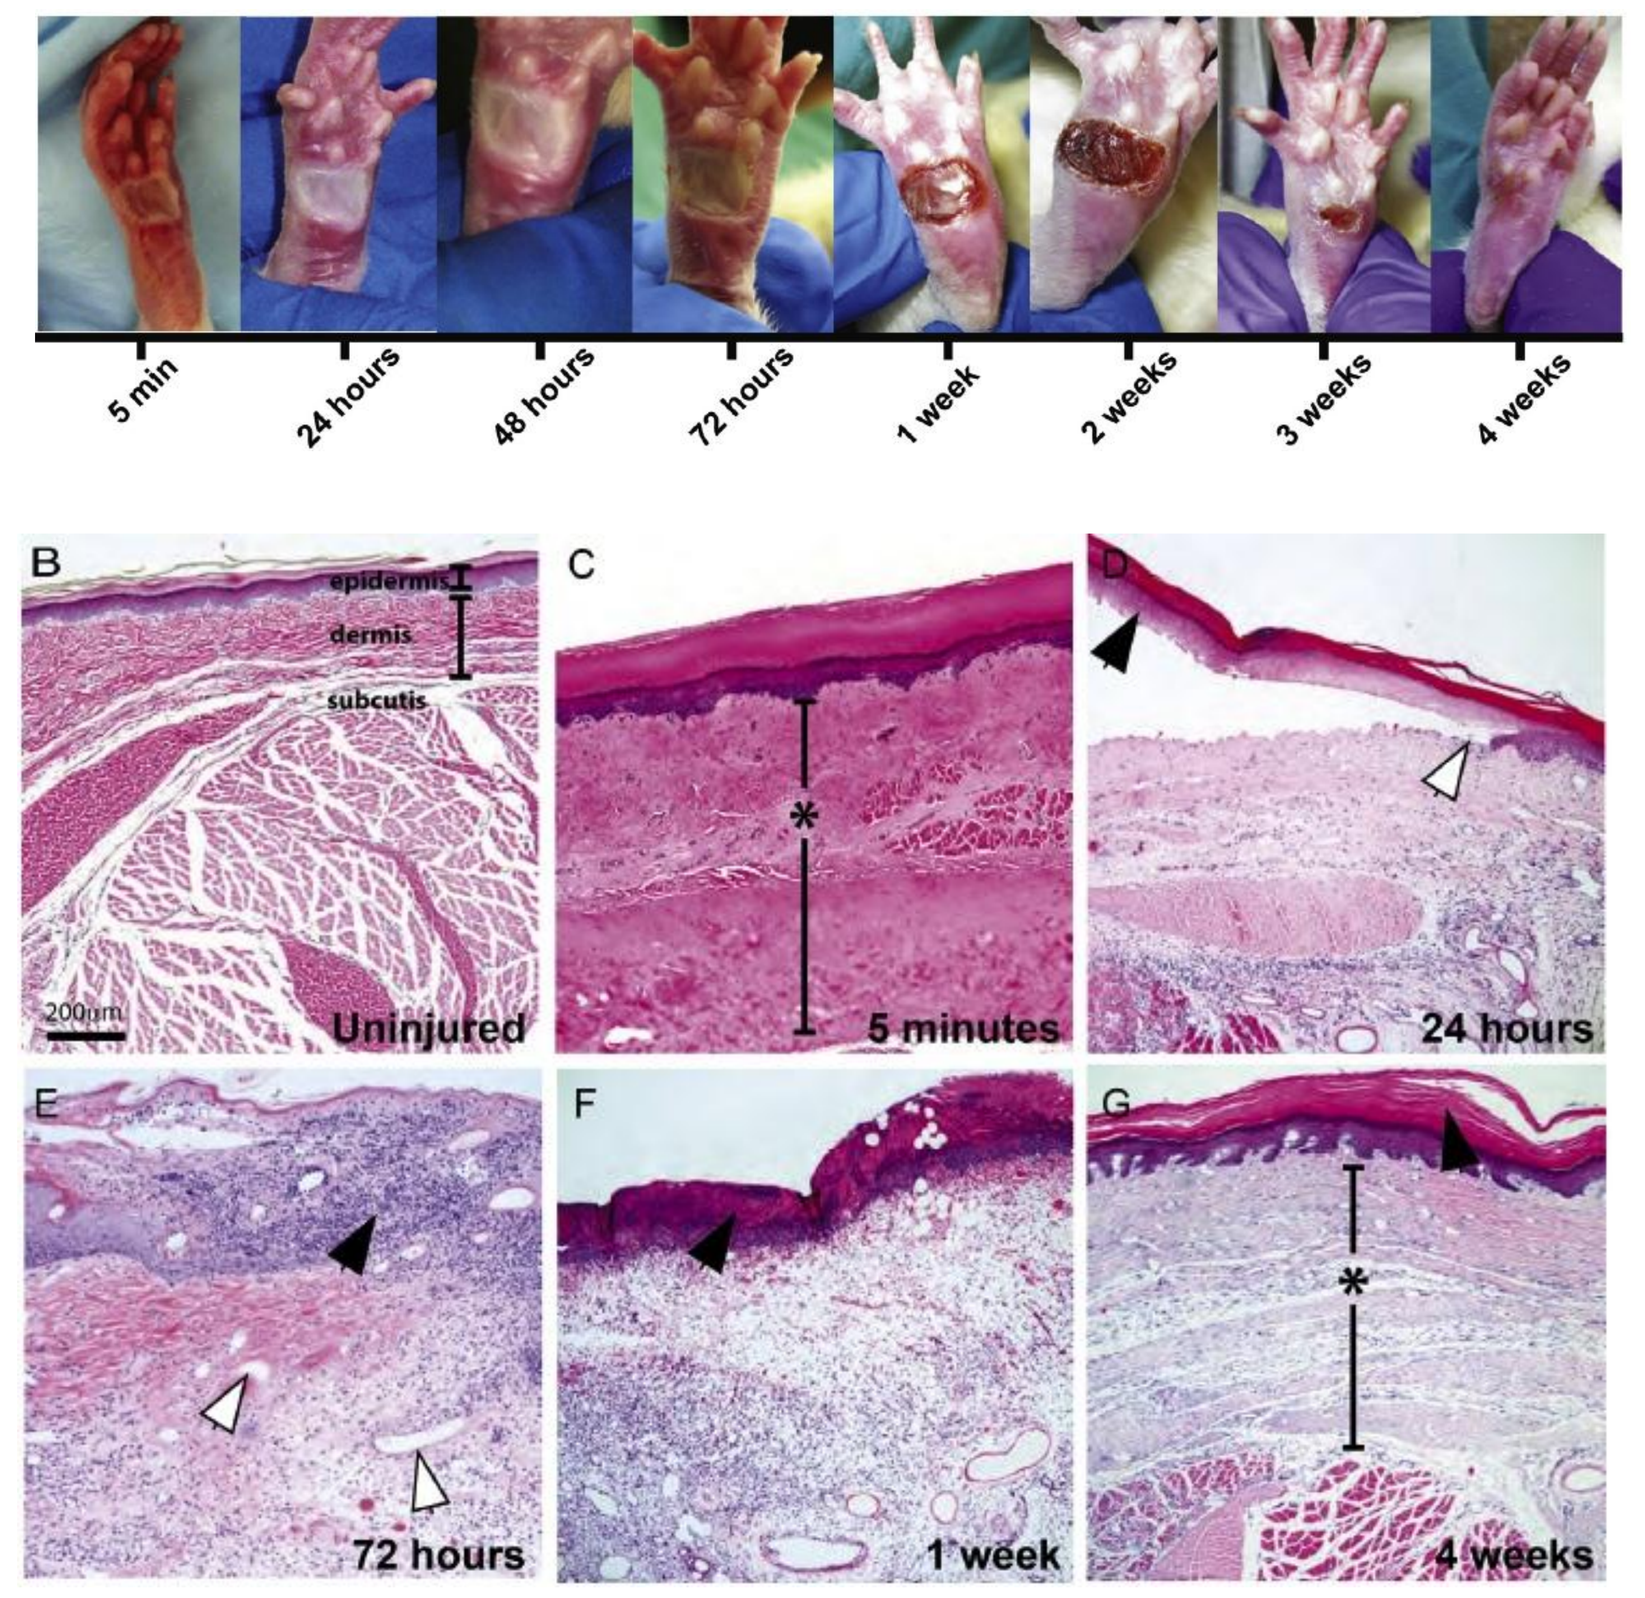
5 min
24 hours
48 hours
72 hours
1 week
2 weeks
3 weeks
4 weeks
B
C
D
E
F
G
epidermis
dermis
subcutis
Uninjured
5 minutes
24 hours
72 hours
1 week
4 weeks

Fowler et al., 2014

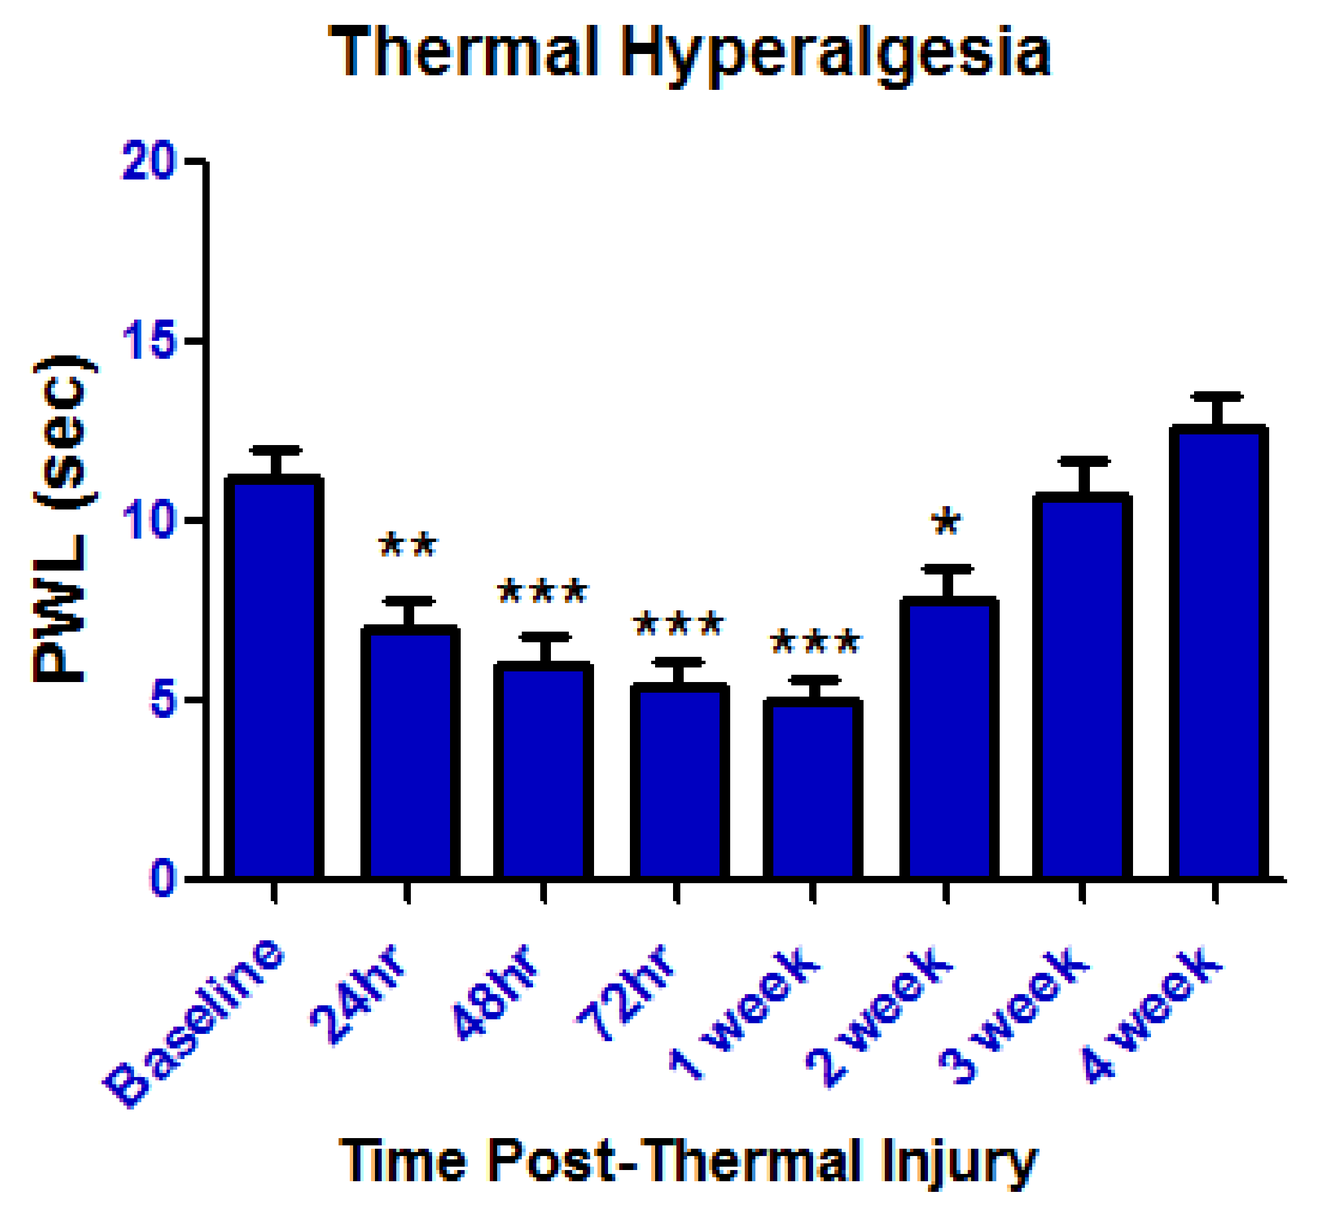
Thermal Hyperalgesia
PWL (sec)
Baseline
24hr
48hr
72hr
1 week
2 week
3 week
4 week
Time Post-Thermal Injury

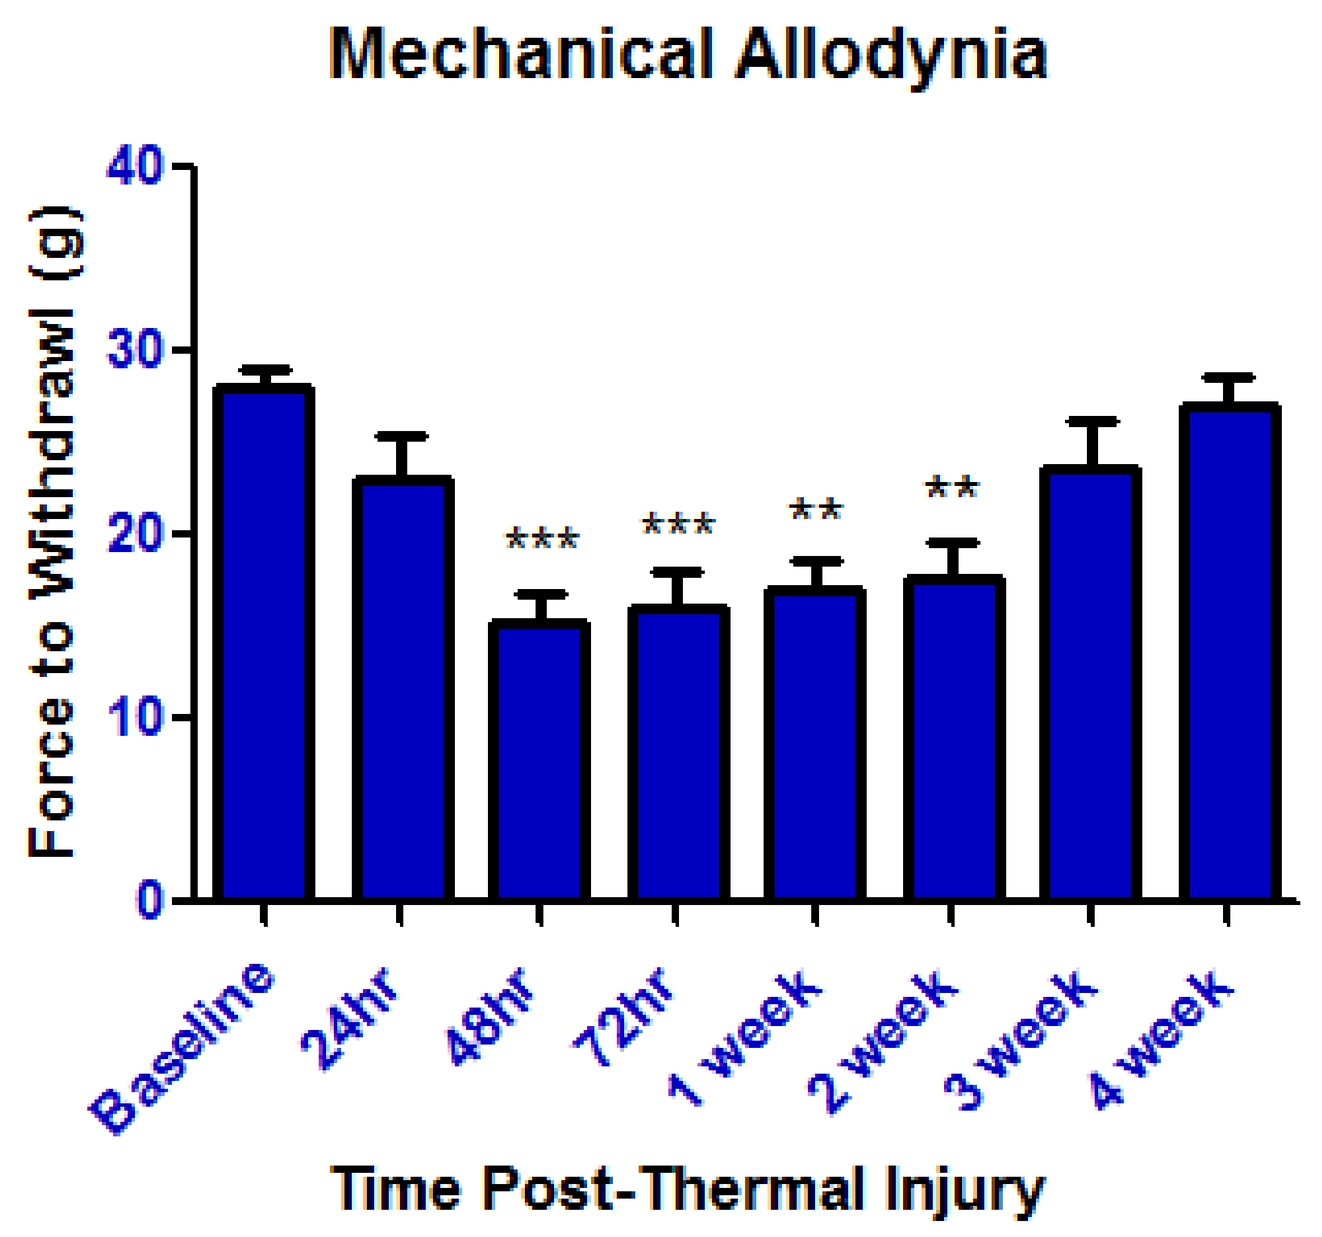
Mechanical Allodynia
Force to Withdrawl (g)
Baseline
24hr
48hr
72hr
1 week
2 week
3 week
4 week
Time Post-Thermal Injury

Tetrodotoxin Attenuates Thermal Hyperalgesia

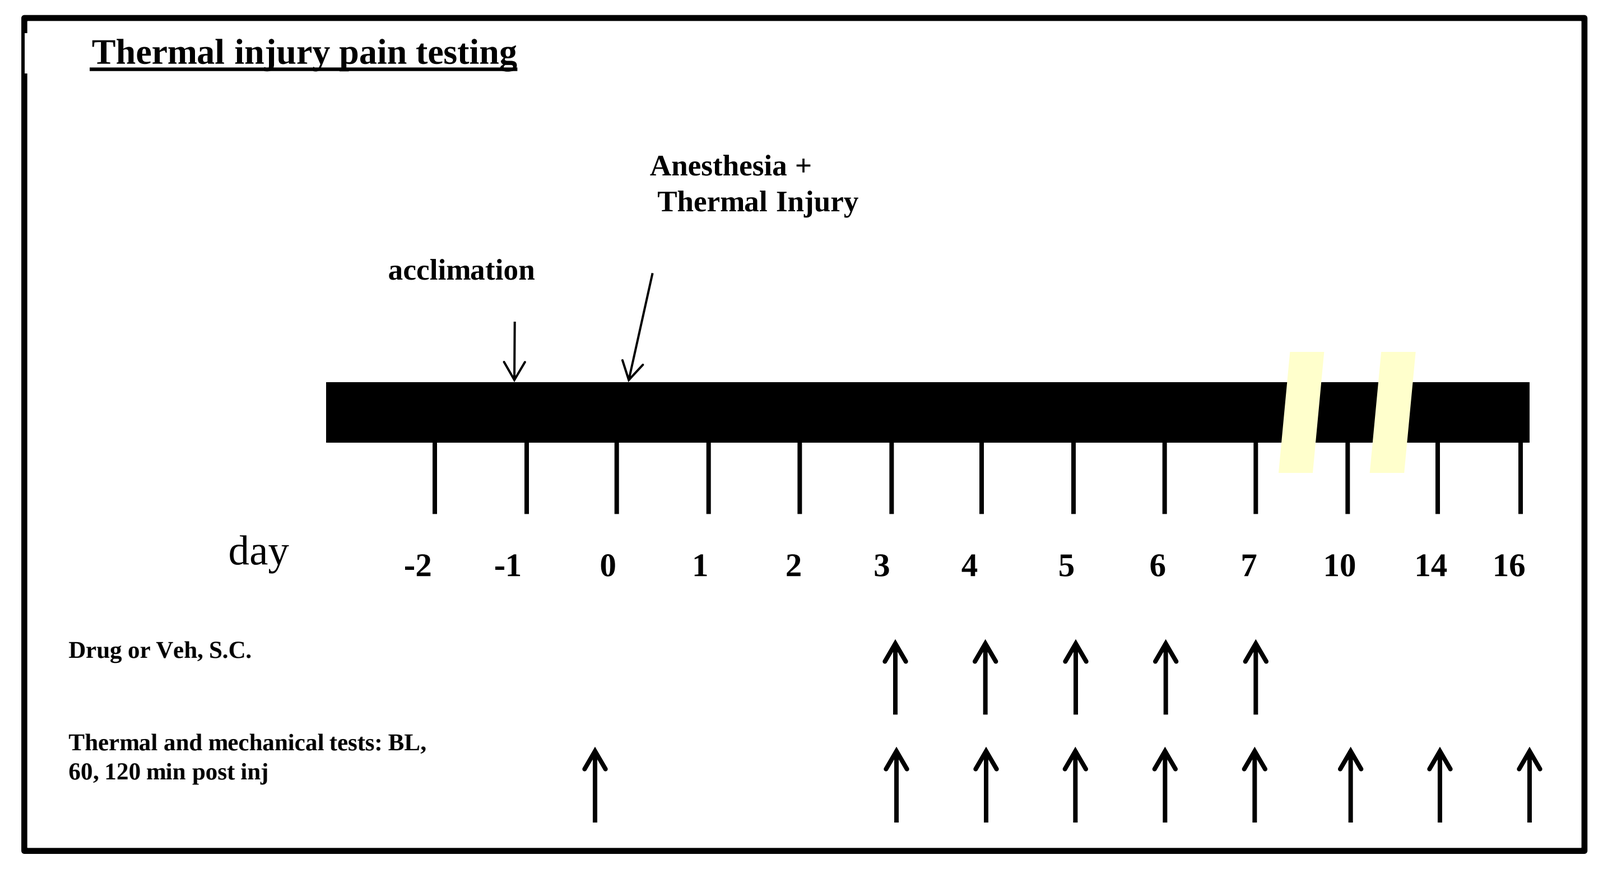
Thermal injury pain testing
Anesthesia + Thermal Injury
acclimation
day
-2 -1 0 1 2 3 4 5 6 7 10 14 16
Drug or Veh, S.C.
Thermal and mechanical tests: BL, 60, 120 min post inj

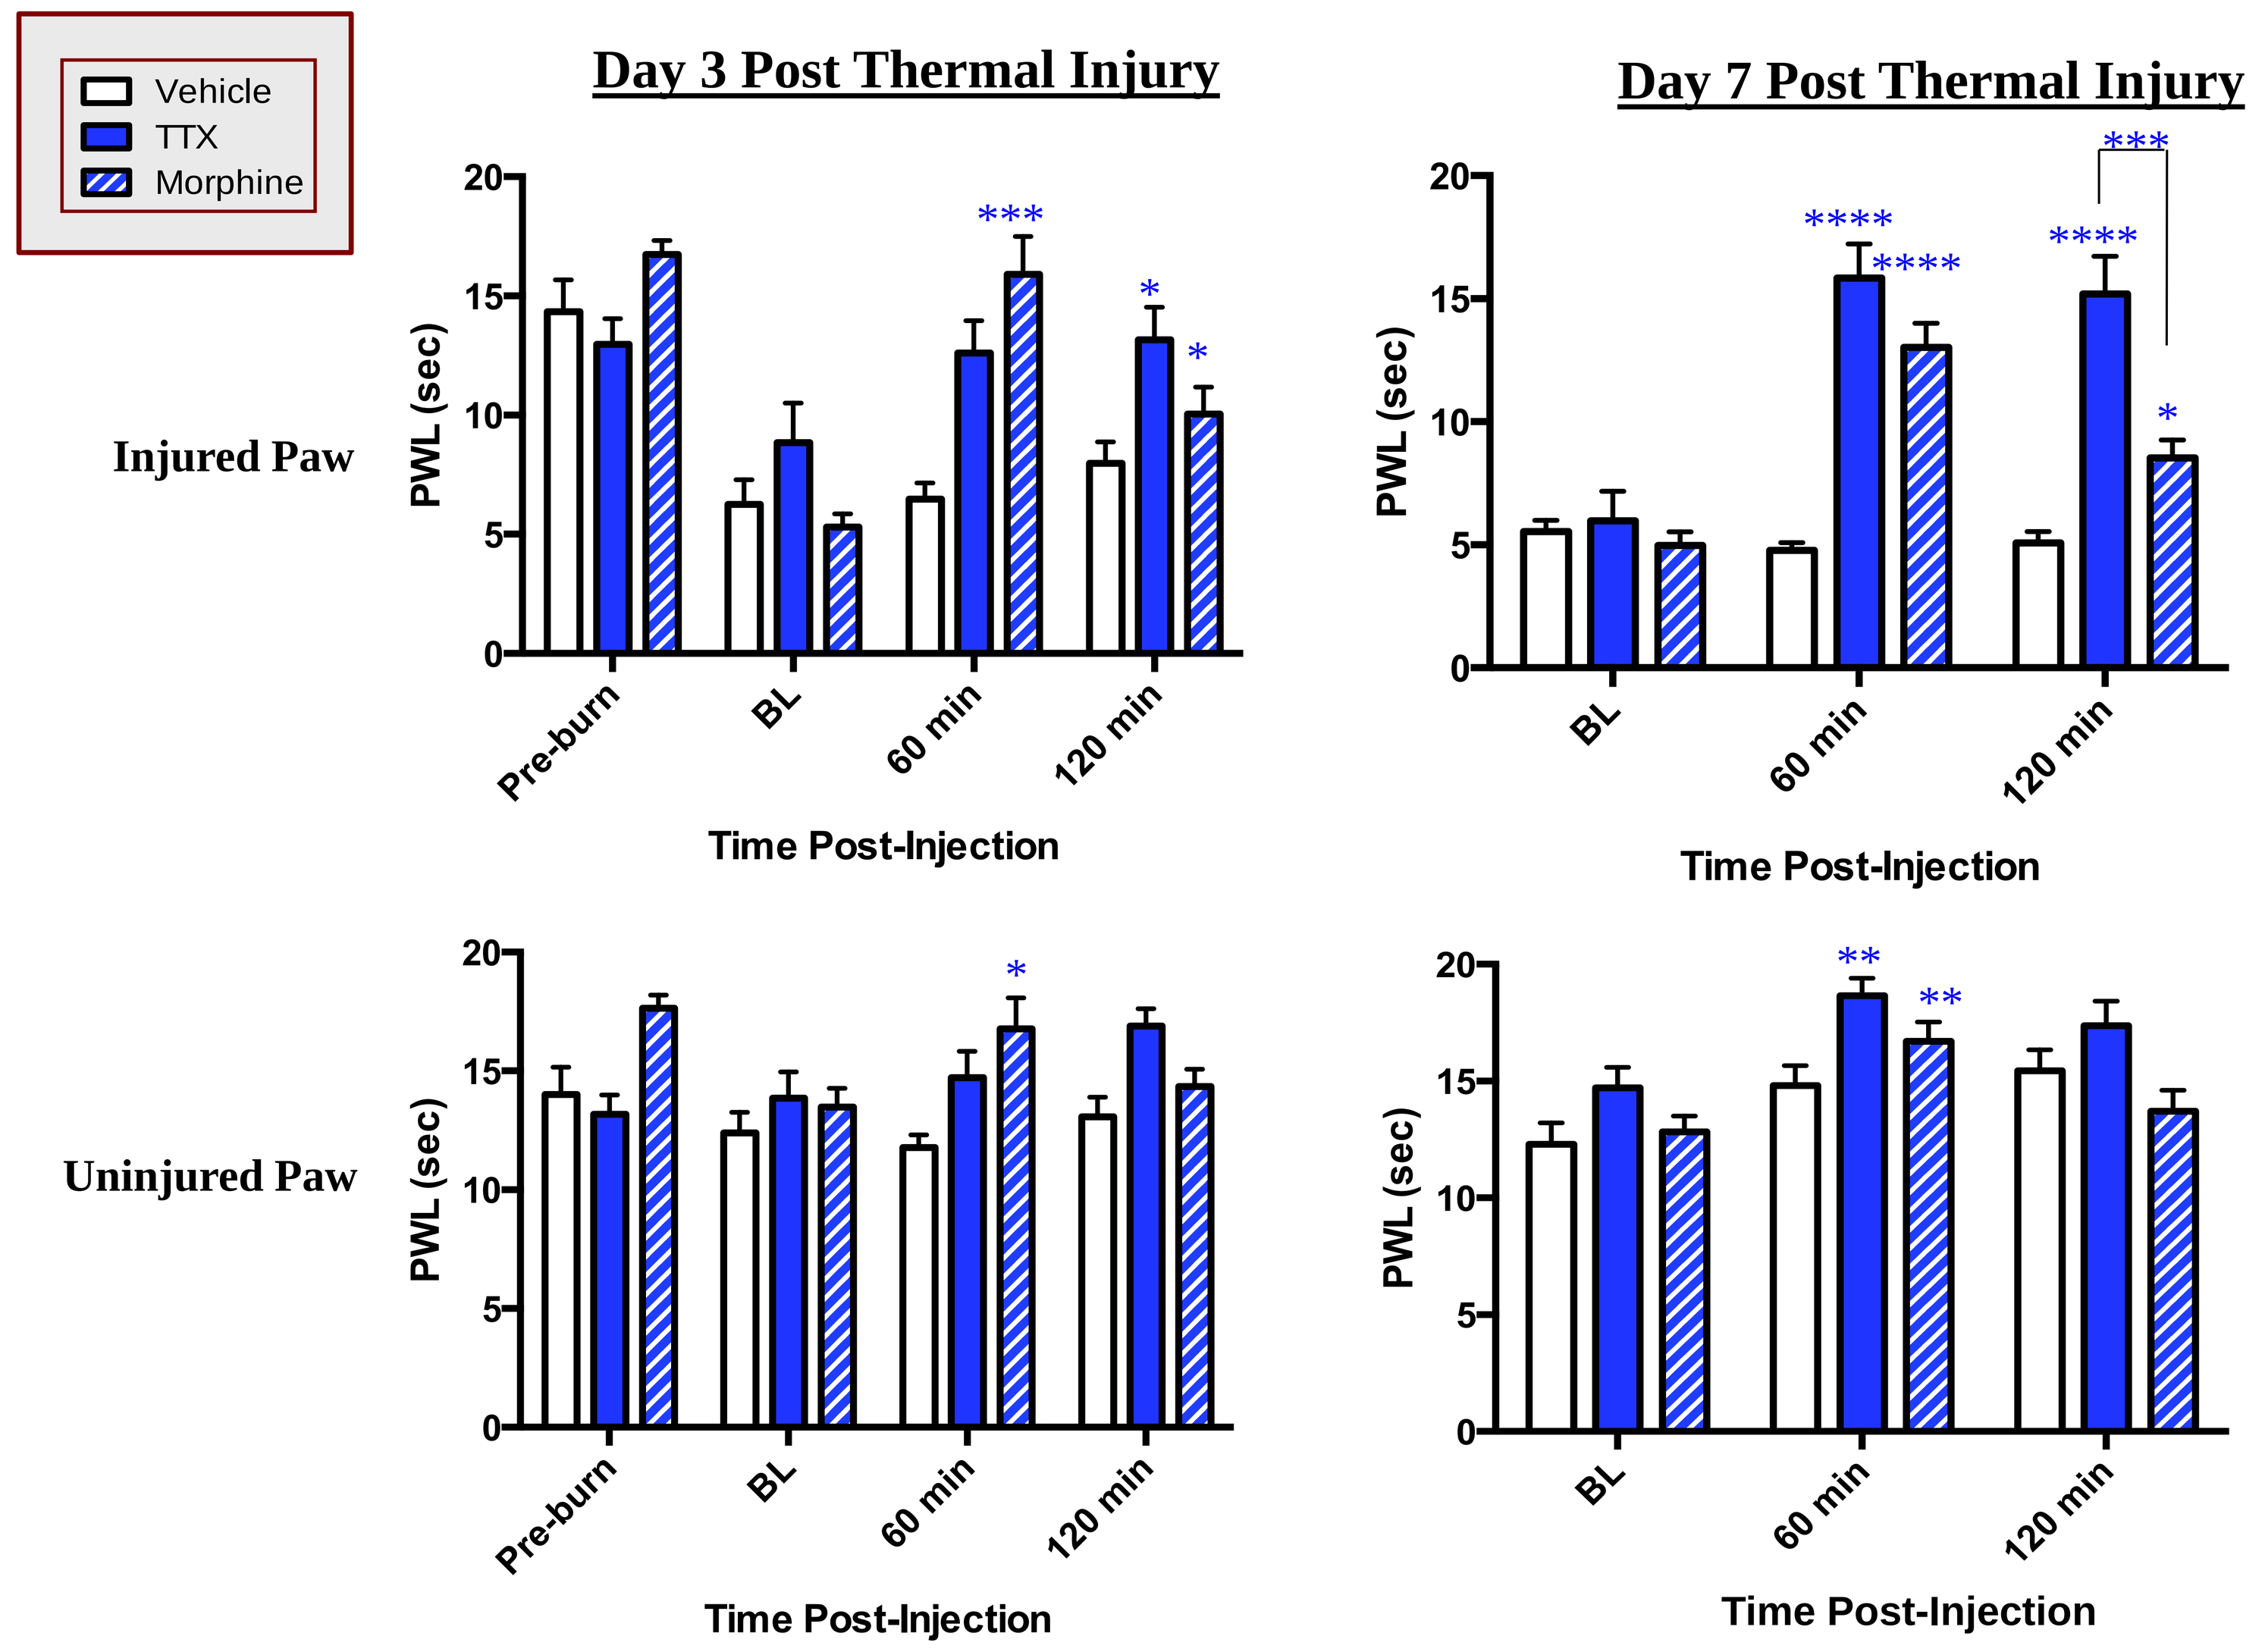
Vehicle
TTX
Morphine
Day 3 Post Thermal Injury
Day 7 Post Thermal Injury
Injured Paw
Uninjured Paw
PWL (sec)
Pre-burn
BL
60 min
120 min
Time Post-Injection

* p<0.05, ** p<0.01, *** p<0.001, **** p<0.0001

Tetrodotoxin Attenuates Mechanical Allodynia

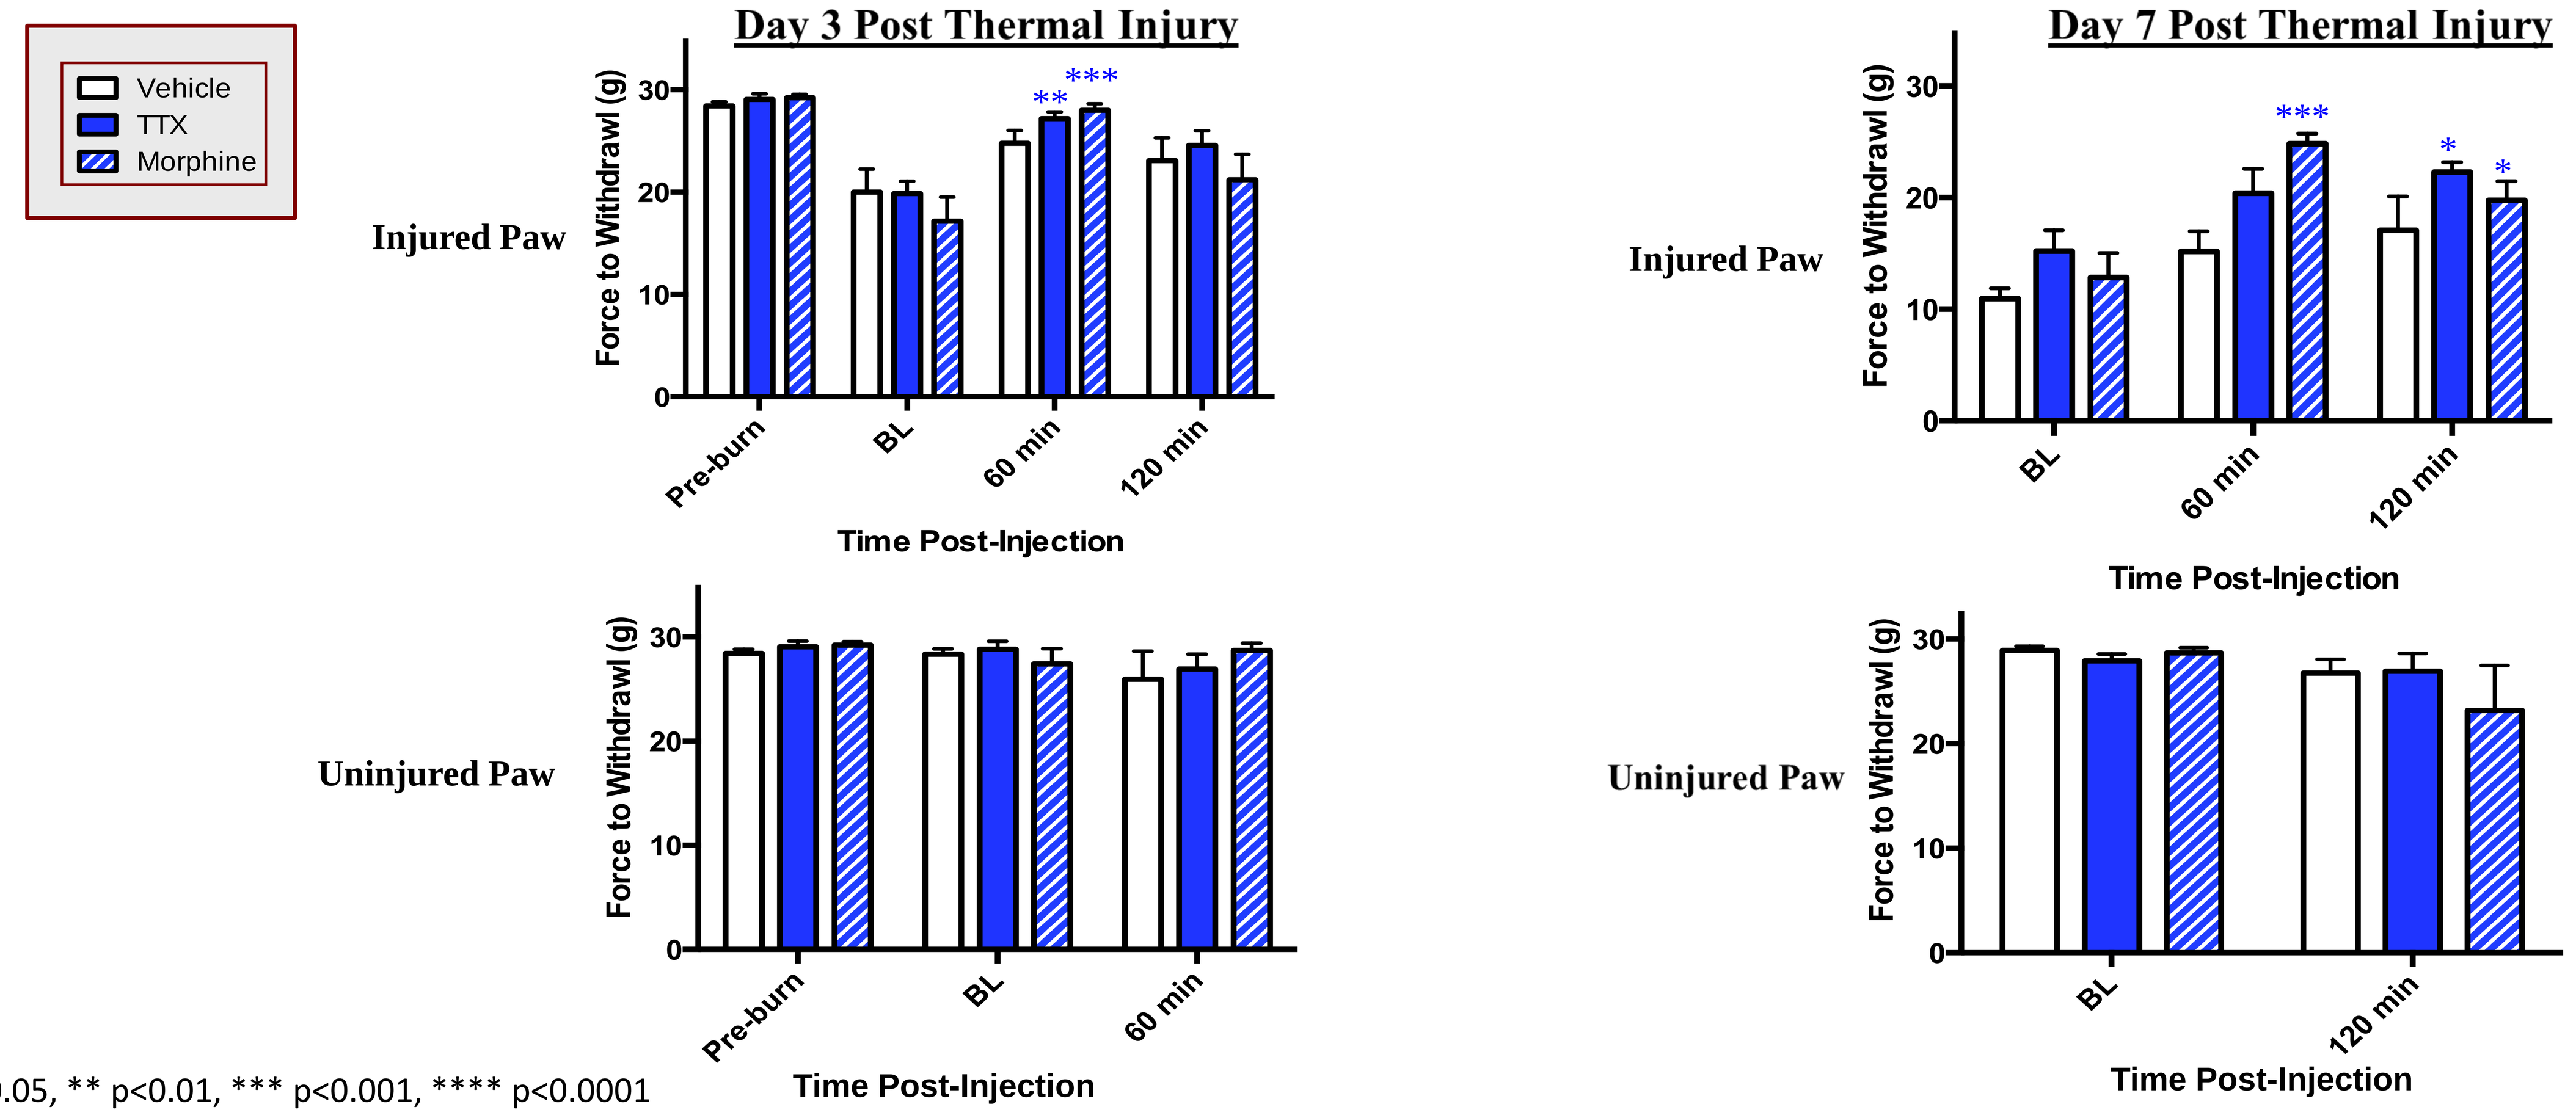
Vehicle
TTX
Morphine
Day 3 Post Thermal Injury
Day 7 Post Thermal Injury
Injured Paw
Uninjured Paw
Force to Withdrawl (g)
Pre-burn
BL
60 min
120 min
Time Post-Injection

* p<0.05, ** p<0.01, *** p<0.001, **** p<0.0001

No Long-Term Effects of TTX Administration in Attenuating Pain Behaviors

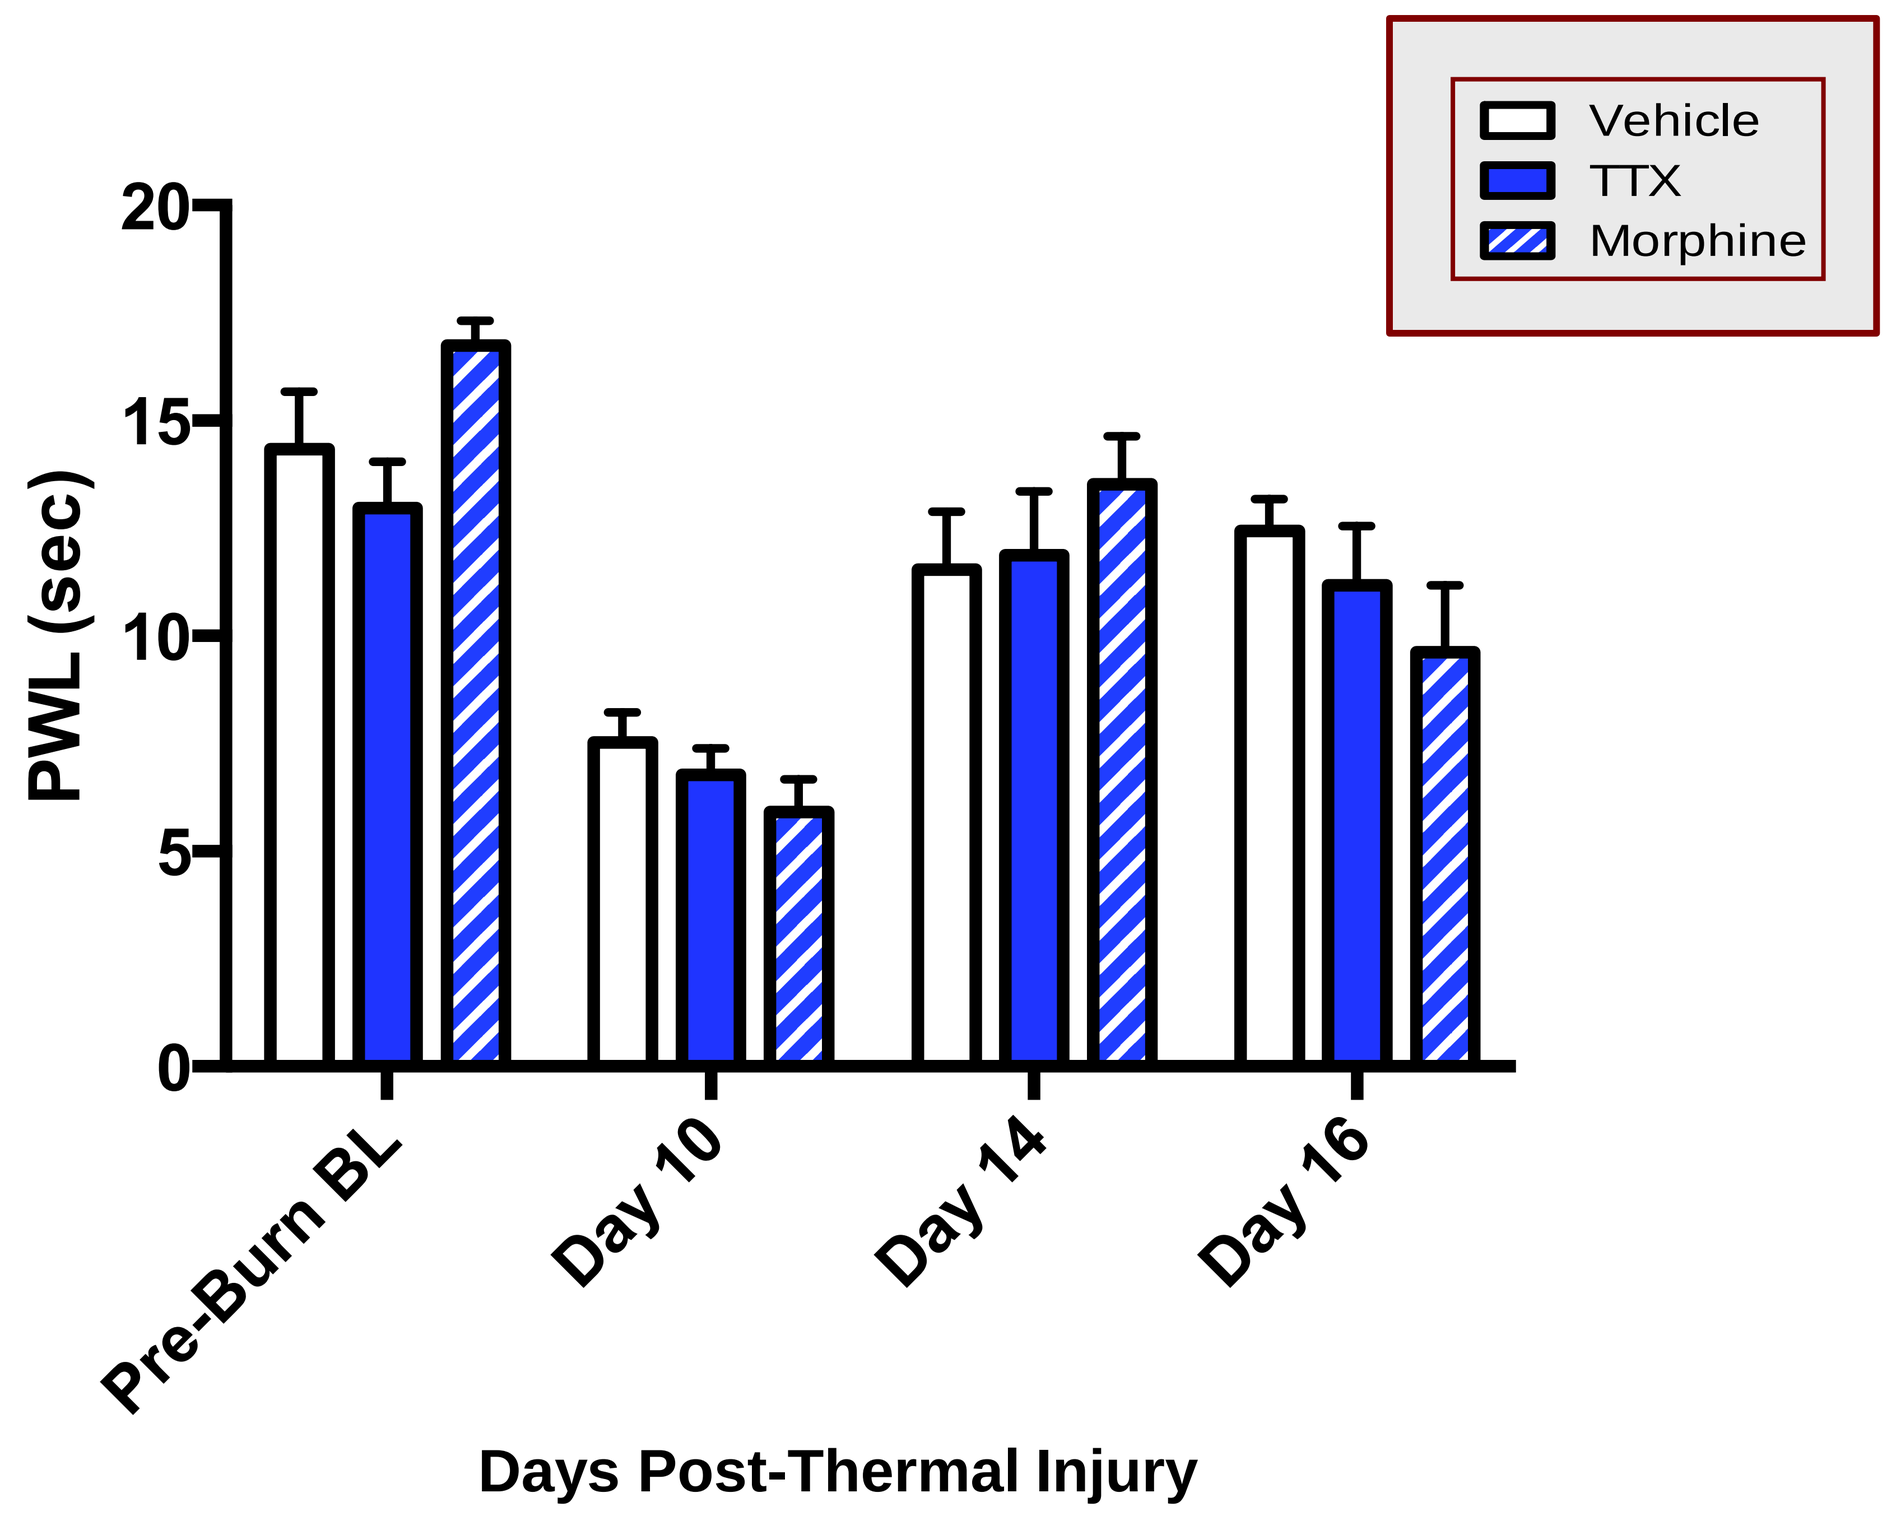
Vehicle
TTX
Morphine
PWL (sec)
Pre-Burn BL
Day 10
Day 14
Day 16
Days Post-Thermal Injury

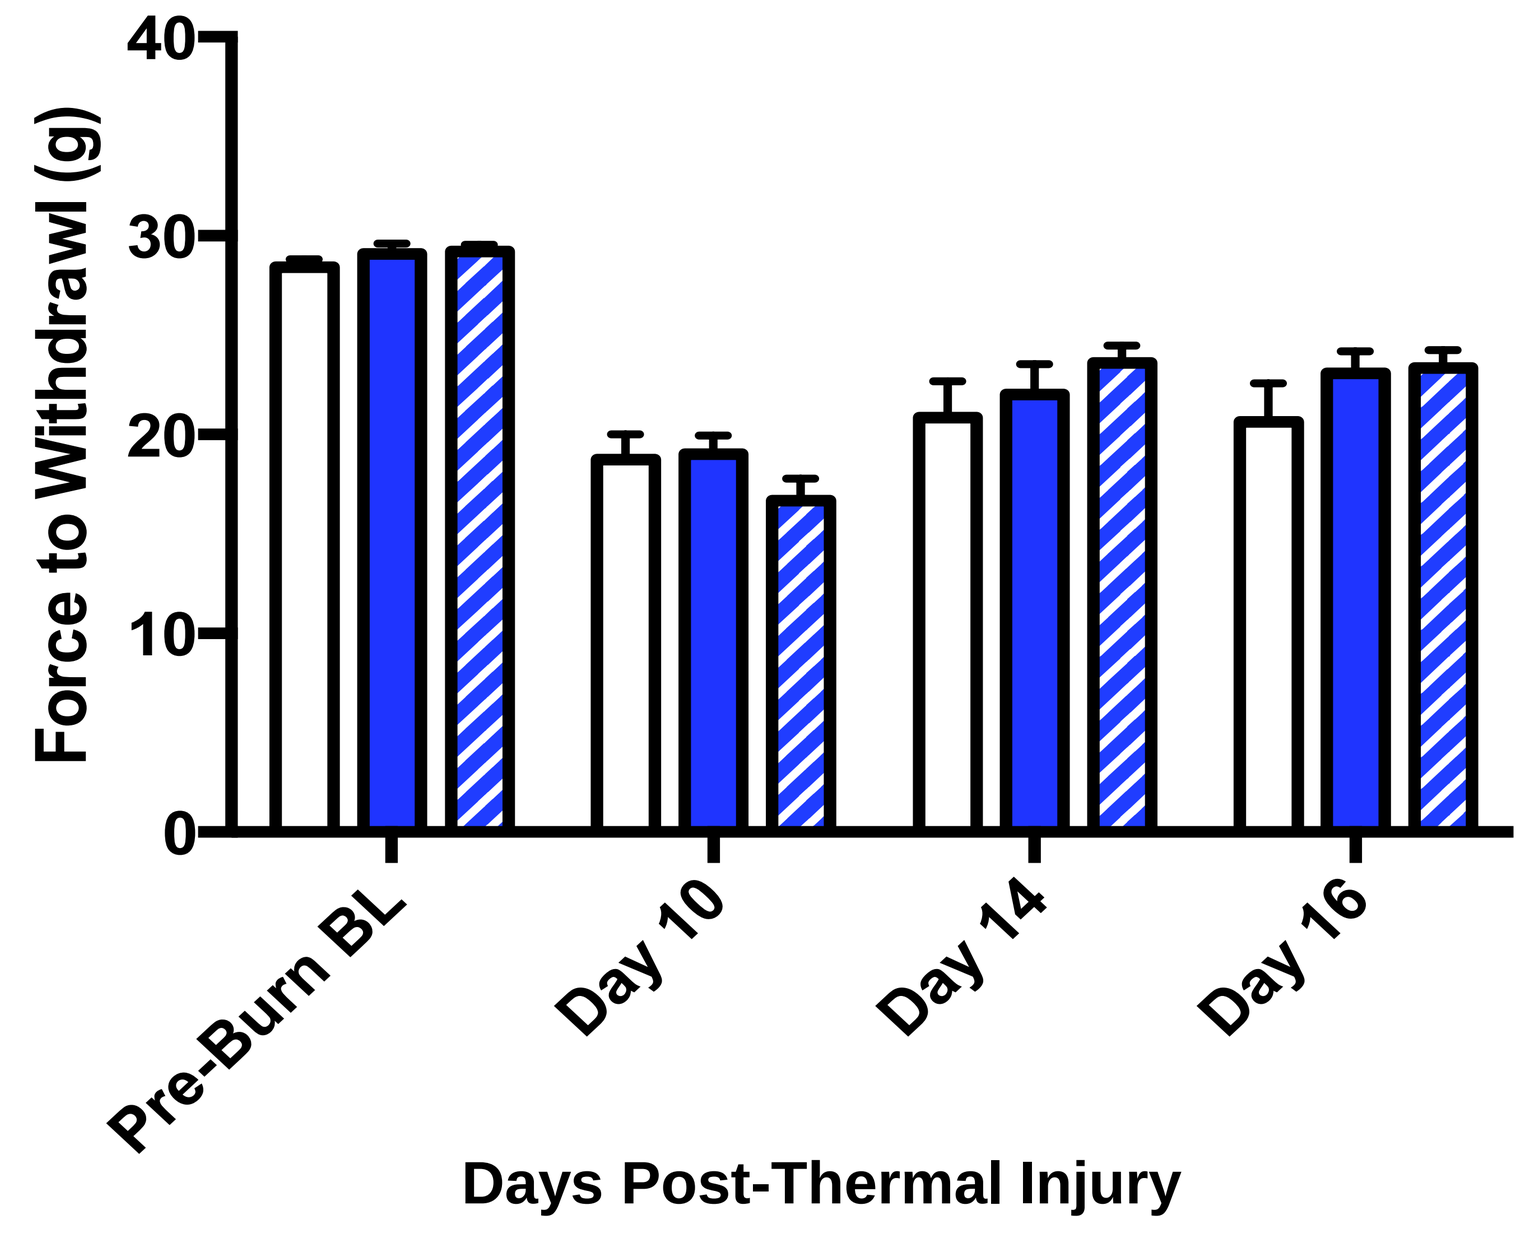
Force to Withdrawl (g)
Pre-Burn BL
Day 10
Day 14
Day 16
Days Post-Thermal Injury

Conclusions

1. Thermal injury was characterized as full thickness based on pathological evidence of the extent of denatured collagen extending into the subcutis.
2. Rats with full thickness thermal injury develop thermal hyperalgesia and mechanical allodynia within 48 hours of injury that lasts up to 2 weeks.
3. TTX significantly reduced thermal hyperalgesia at all-time points tested.
4. Repeated doses of TTX significantly reduced thermal hyperalgesia as compared to repeated doses of morphine.
5. TTX reduced mechanical allodynia and was similar to morphine.

Clinical Significance: These results indicate that blocking TTX sensitive sodium channels with low concentration of TTX may dampen pain detection during evacuation of Wounded Warriors and throughout painful procedures of burn wound care/healing. Also, the results indicate TTX may have a reduced risk of the development of tolerance after repeated doses as compared to morphine.

Acknowledgements

The opinions and assertions contained herein are the private views of the authors and are not to be construed as official or as reflecting the views of the Department of the Army or the Department of Defense.

This study has been conducted in compliance with the Animal Welfare Act, the implementing Animal Welfare Regulations, and the principles of the Guide for the Care and Use of Laboratory Animals.

This work was funded by the U.S. Army Institute of Surgical Research and the Division of Intramural Research, NIDCR, NIH. Dr. Salas is the recipient of a National Research Council Fellowship. The authors would like to acknowledge Combat Casualty Care as well as Rehabilitative Medicine.

This work was accomplished in collaboration with WEX Pharmaceuticals [2]